Poster # P.29

Accelerating clinical development of new therapeutics with patient data: evidence from the collaborative Trajectory Analysis Program (cTAP) in DMD

E. Mercuri[1], C. McDonald[2], SJ Ward[3], J. Signorovitch [3],[4], F. Muntoni[5], N. Goemans [6], B. Wong [7], K. Vandenborne [8], A. Manzur [5] and G. Sajeev[4]

[1] Universitario "A. Gemelli" Largo A. Gemelli 8 00168 Roma, Italy [2] Department of Physical Medicine and Rehabilitation, and Pediatrics, University of California, Davis - Davis (United States)
[3] collaborative Trajectory Analysis Project - Cambridge (United States) [4] Analysis Group, Inc. - Boston (United States) [5] Dubowitz Neuromuscular Centre, NIHR Great Ormond Street Hospital Biomedical Research Centre, Great Ormond Street Institute of Child Health, University College London, & Great Ormond Street Hospital Trust - London (United Kingdom)
[6] University Hospitals Leuven, Child Neurology - Leuven (Belgium) [7] Department of Pediatrics, University of Massachusetts Medical School, Worcester, MA 01655, USA
[8] Department of Physical Therapy, University of Florida, Gainesville, FL, US

Background

- Patients DMD and their families generously consent to research on their clinical data in the hope of accelerating potential new treatments
- Although it is widely believed that curated patient data are invaluable to drug development, the published literature is sparse on *how* patient data are used

Study Objectives

- Characterize how research on anonymized patient data aggregated by cTAP is used to accelerate clinical development of new therapeutics
- Provide clinical experts and developers with a framework for sharing the importance of curated patient data in drug development

Methods

Figure 1. Anonymized patient data

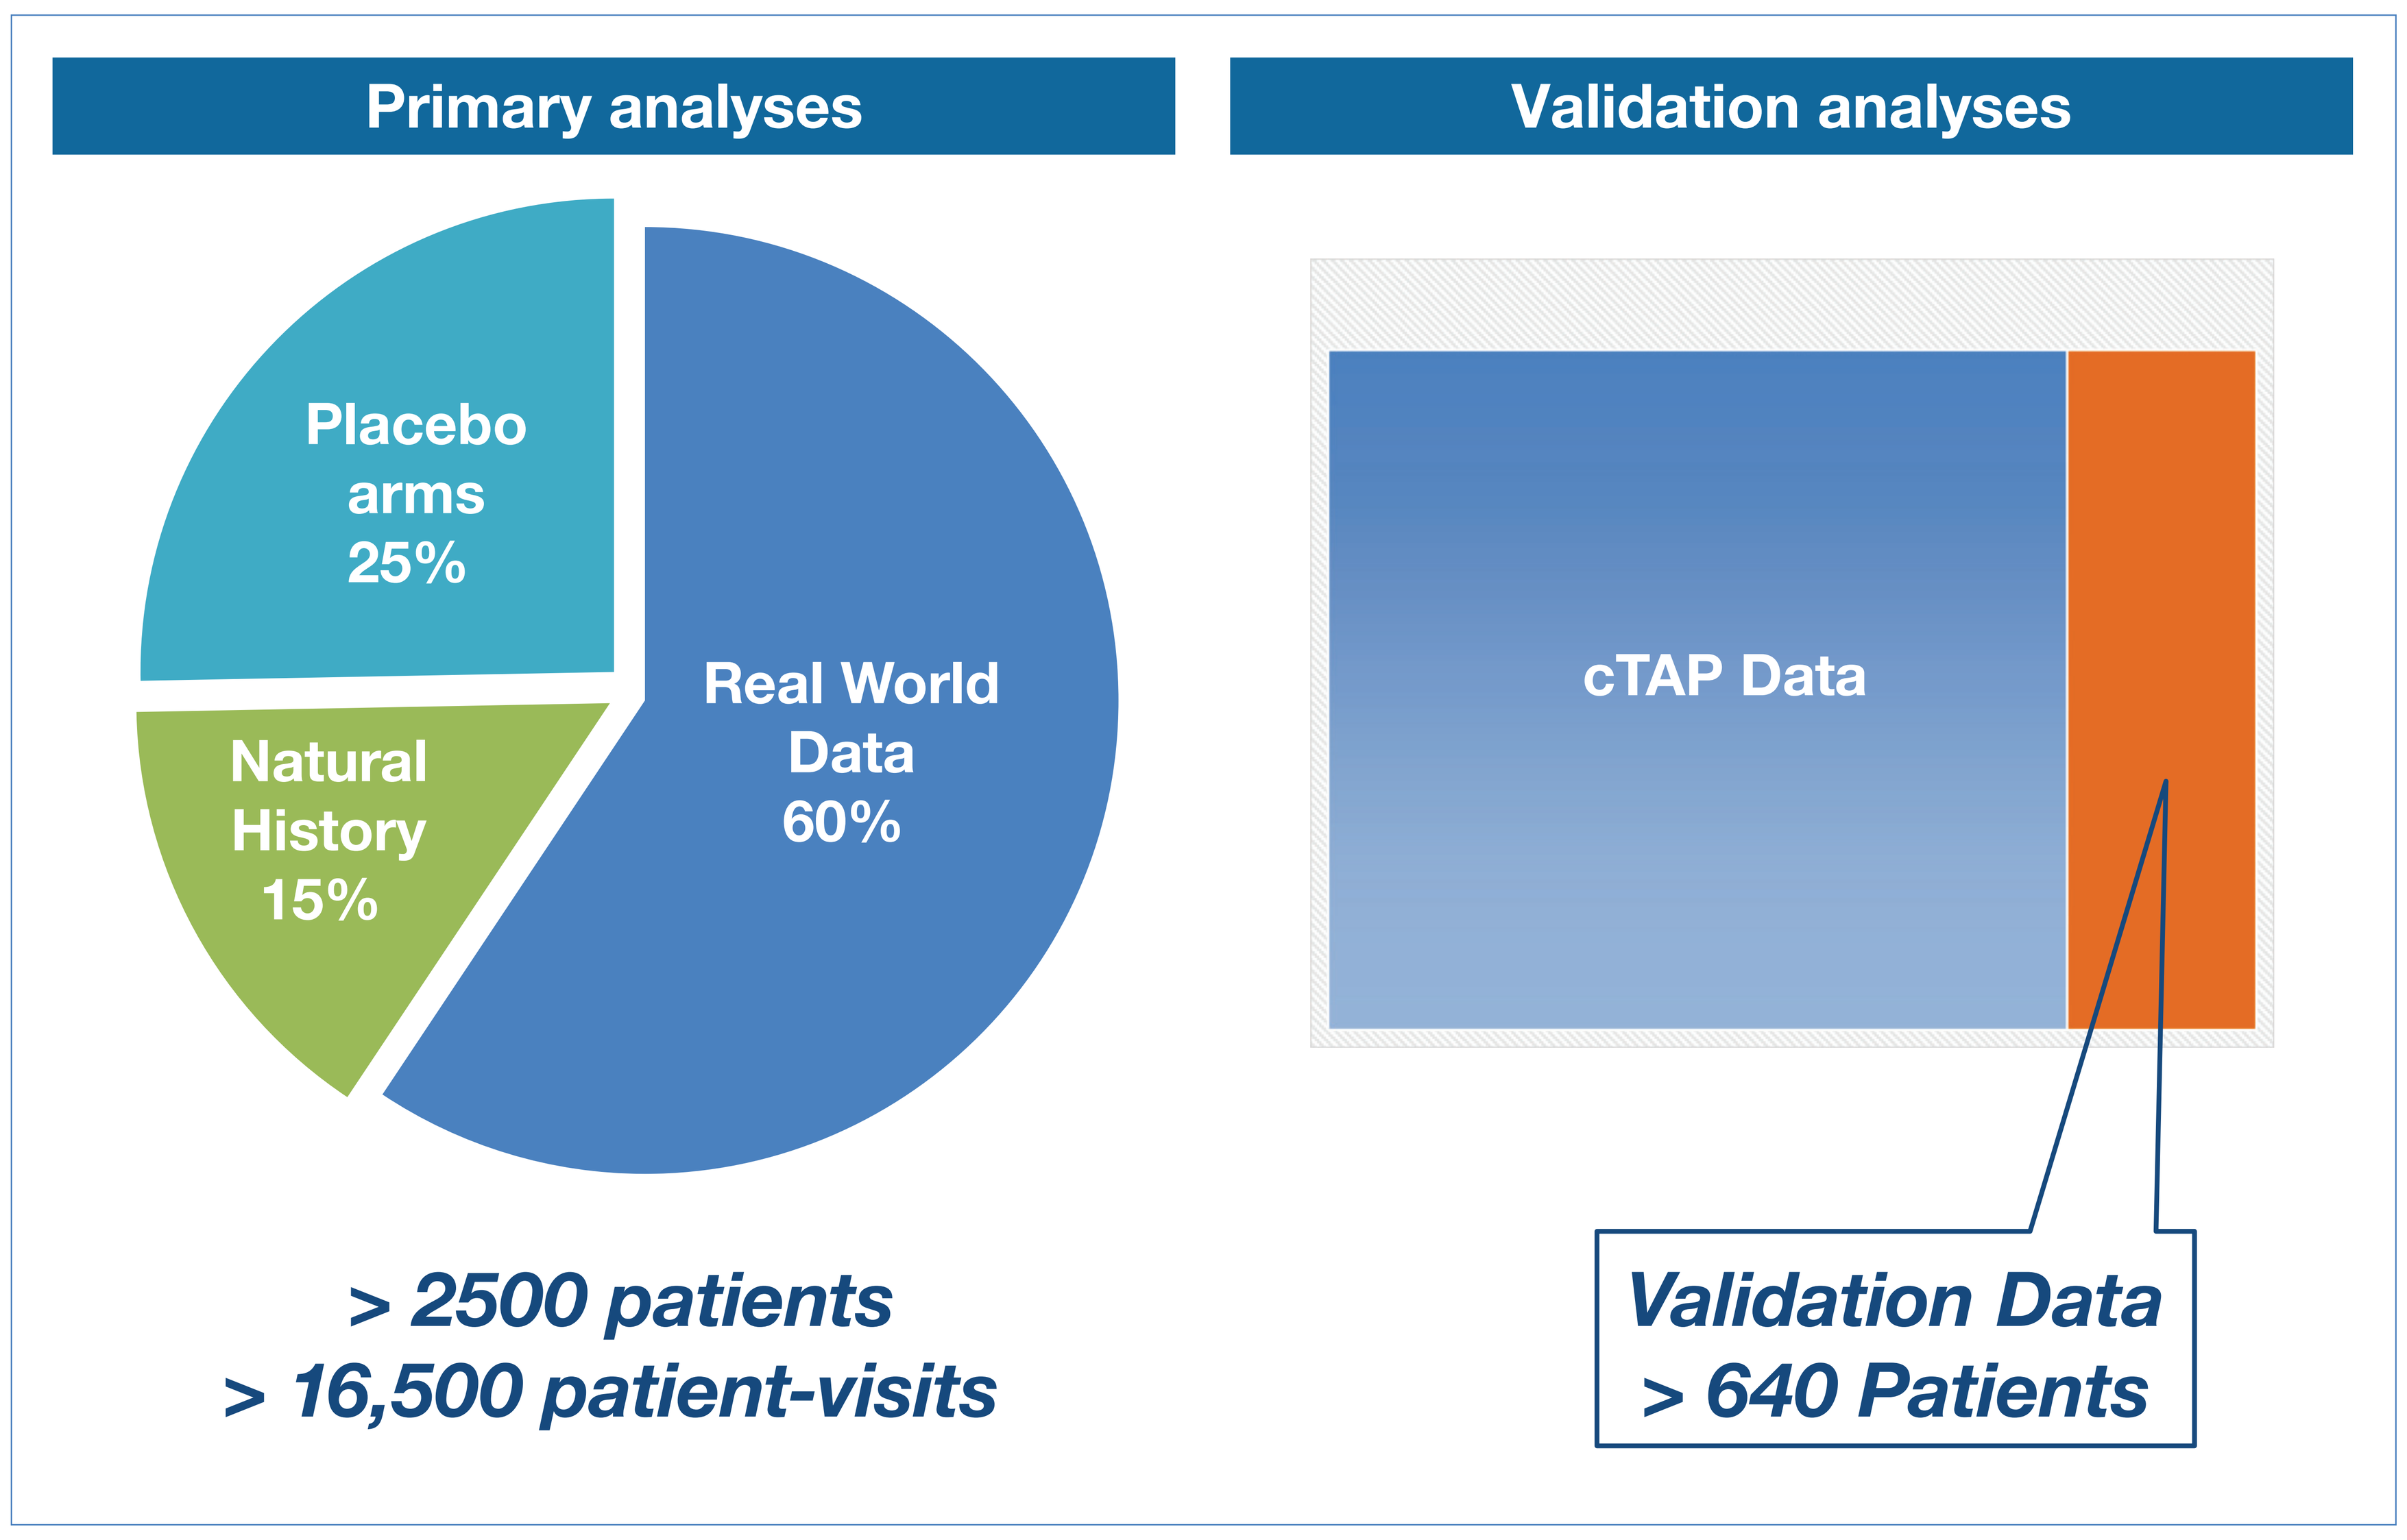
Primary analyses
Validation analyses
Placebo arms 25%
Natural History 15%
Real World Data 60%
cTAP Data
> 2500 patients
> 16,500 patient-visits
Validation Data
> 640 Patients

Figure 2. Categorizing the purposes of analyses of patient data

Drug Development Aims	Analytic Objectives
Learn how the data behave	▪ Characterize longitudinal trajectories ▪ Assess variability of clinical endpoints ▪ Identify prognostic factors
Design and power a clinical trial	▪ Functional trajectories of patients that meet trial Inclusion/exclusion criteria ▪ Define clinically meaningful progression
Analyze and interpret results of a clinical trial	▪ Adjust for prognostic factors at baseline ▪ Stratify to account for any difference between cohorts in baseline function
Evidence for regulatory approval and access	▪ Match patients enrolled in the drug trial to natural history and prior placebo controls ▪ Model impact of drug on future clinical milestones

Results

- We analyzed the 78 discrete analytical studies conducted 2015 – 2021
- Approximately 65% of studies were conducted within the cTAP pre-competitive collaboration and approximately 35% applied these collaborative findings to company-specific clinical trials
- To date the majority of studies (43%) have focused on helping drug companies to optimize design and power of clinical trials (**Figure 3**)

Figure 3: purpose of analyses of patient data 2015-2021

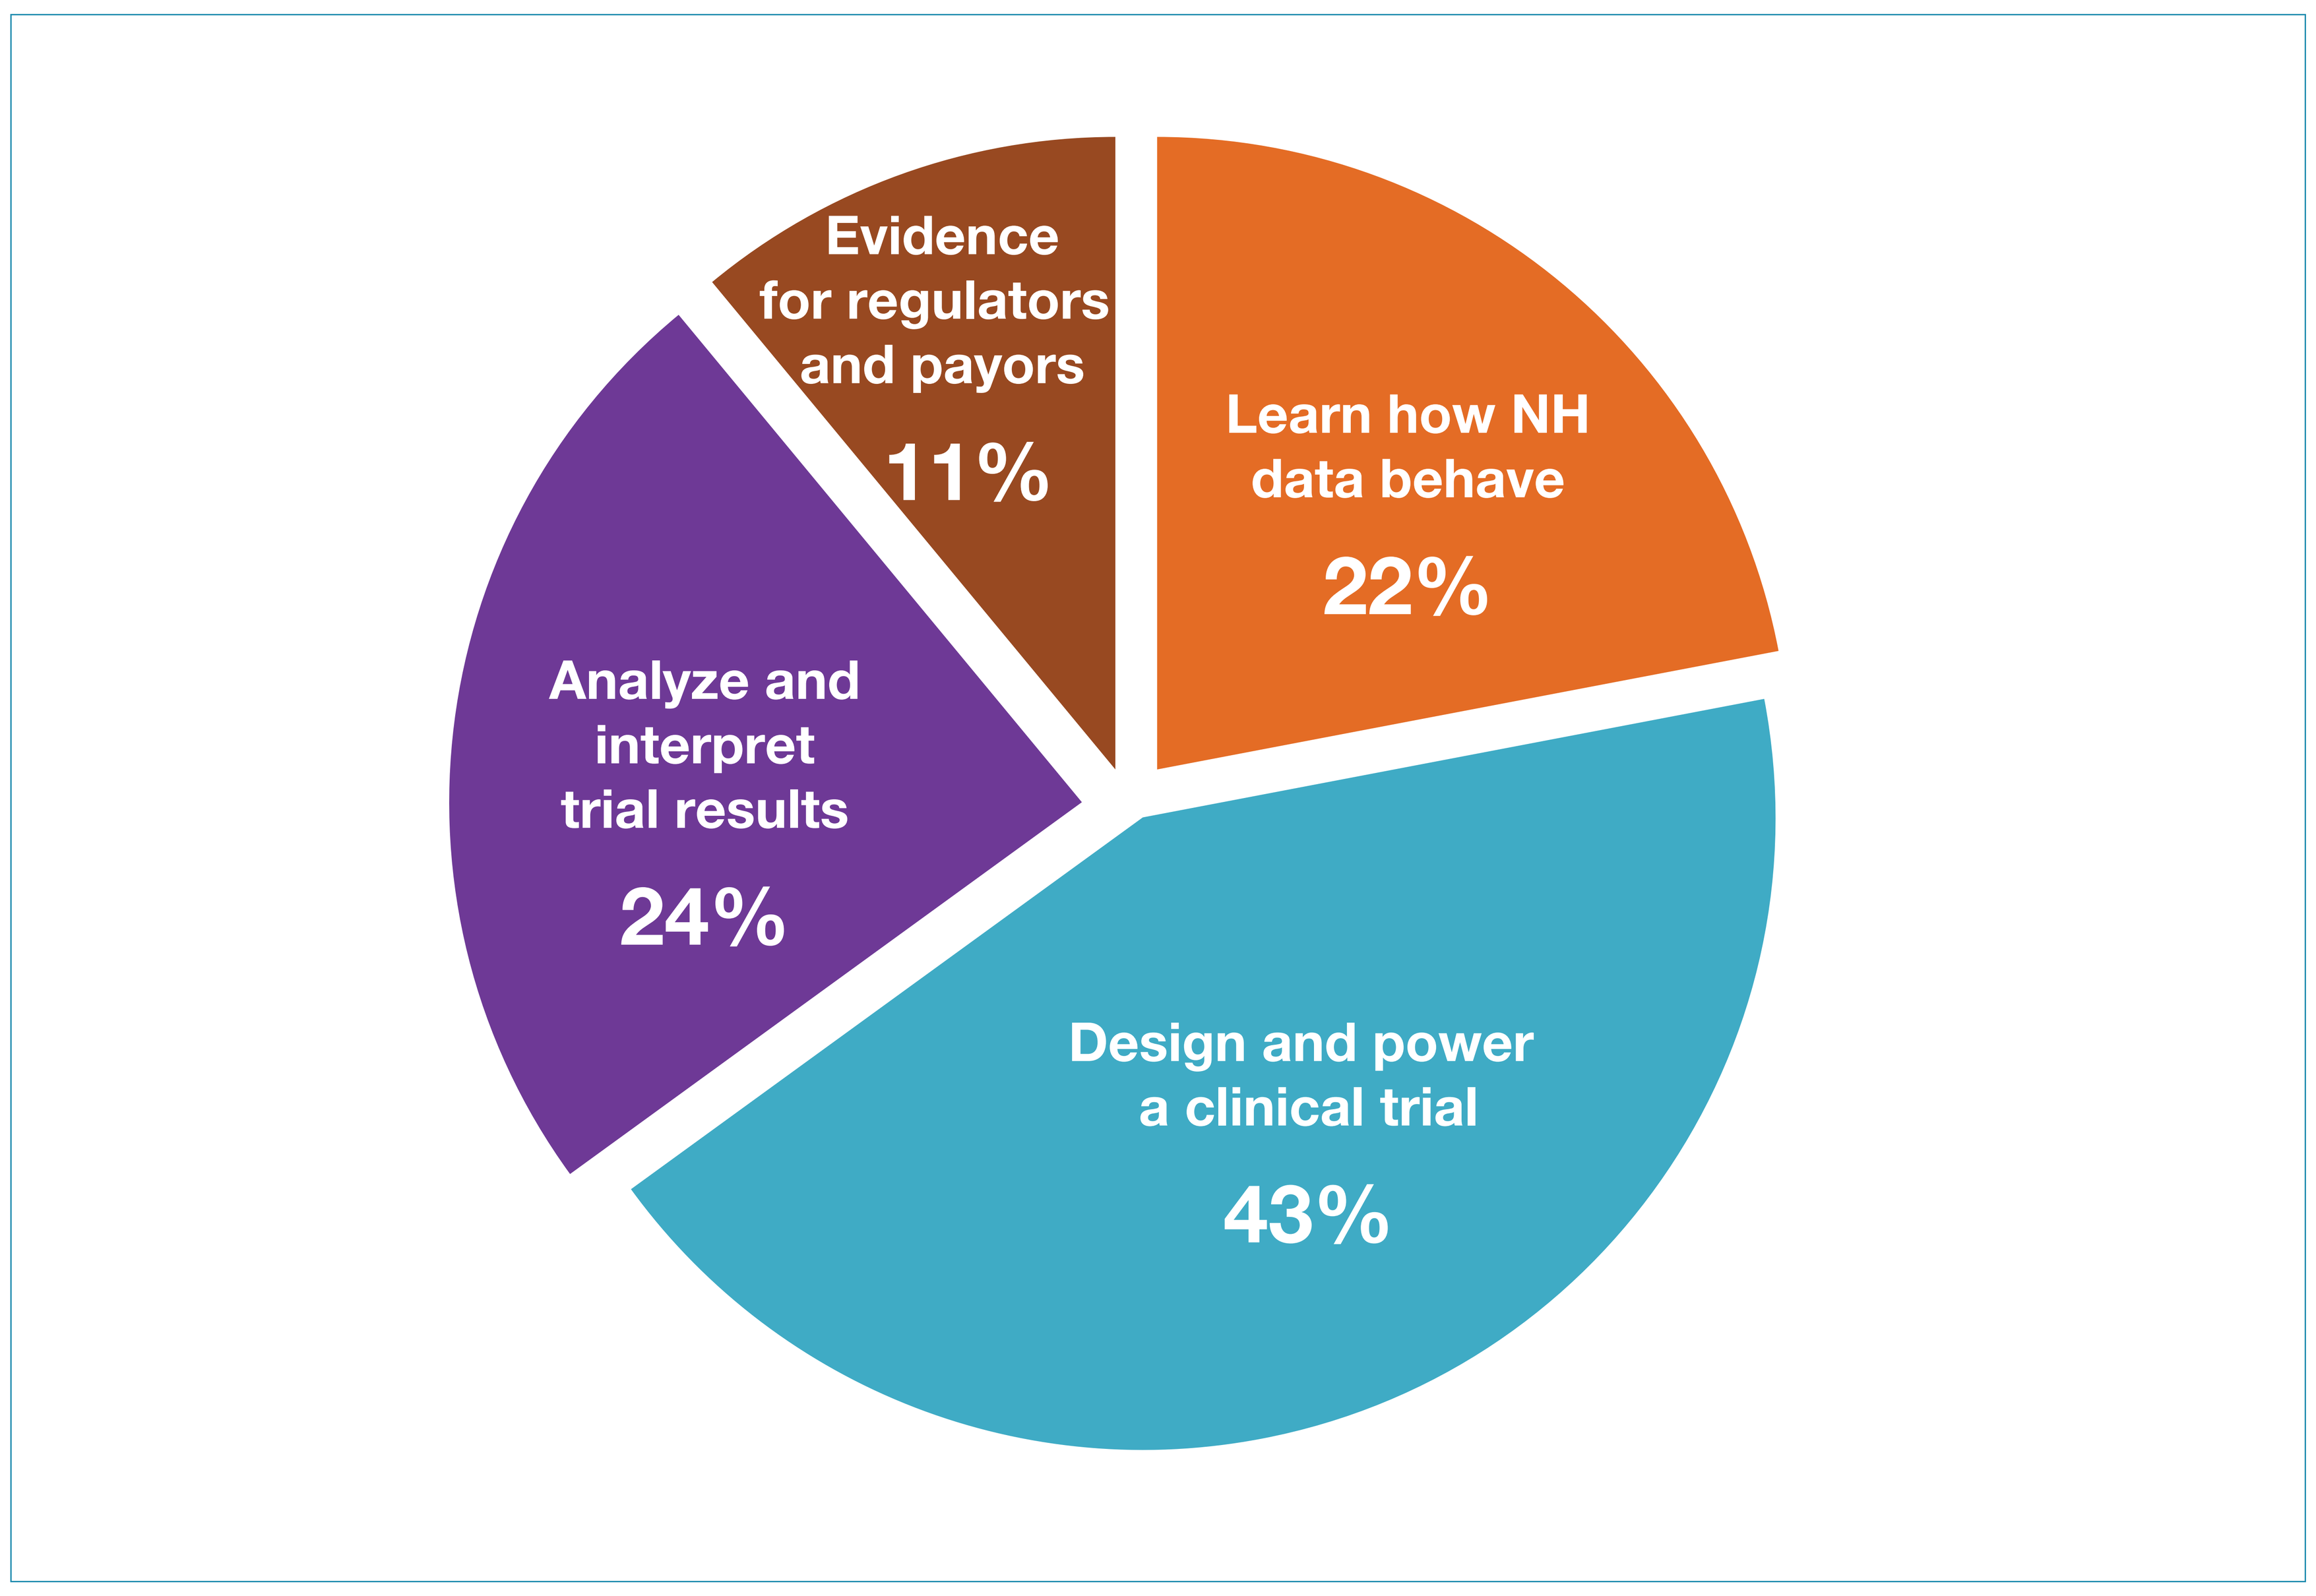
Evidence for regulators and payors 11%
Learn how NH data behave 22%
Analyze and interpret trial results 24%
Design and power a clinical trial 43%

The purpose of cTAP analytical studies on patient data has evolved and expanded over time (Figure 4).

- The purpose of cTAP analytical on patient data has evolved and expanded over time (**Figure 4**).
- Initial studies focused on providing a richer characterization of natural history progression. Such studies continue to be important but have essentially plateaued since 2019 at ~ 20%
- Studies focused on clinical trial design, analysis, and interpretation have grown rapidly year on year since 2016
- In 2019, the first studies to support evidence-based drug effect for regulatory approval and patient access were conducted, and is growing rapidly
- Expansion of data use and evolution of scope parallels the growth in cTAP membership

Figure 4. Drug development focus of analytical studies over time

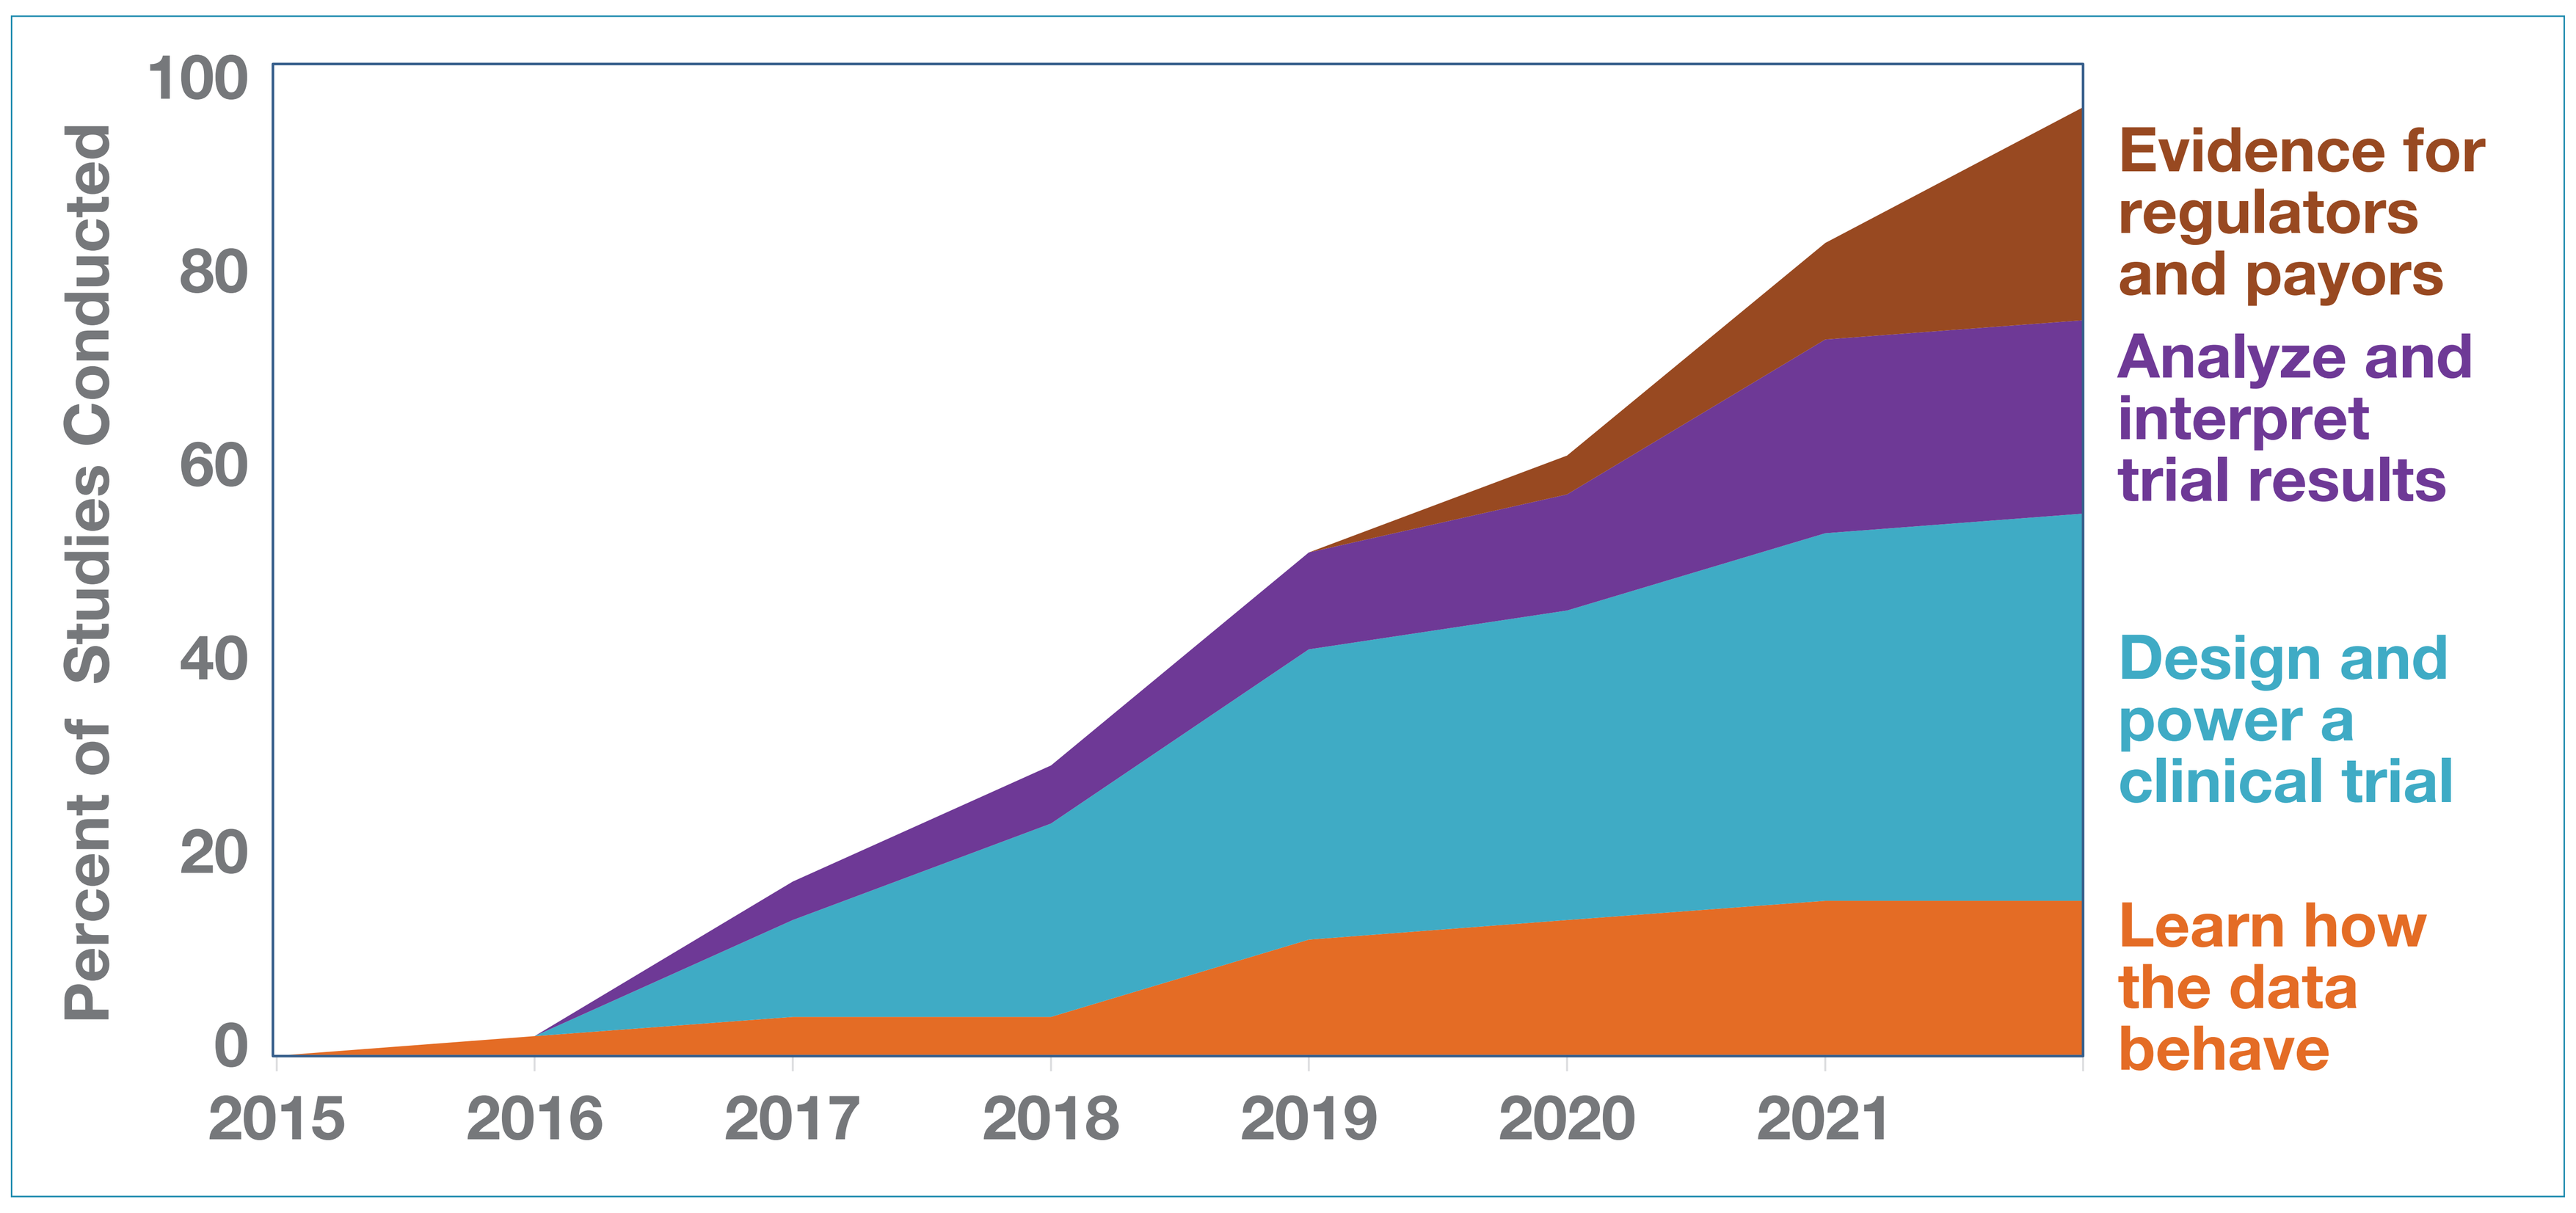
Percent of Studies Conducted
100
80
60
40
20
0
2015
2016
2017
2018
2019
2020
2021
Evidence for regulators and payors
Analyze and interpret trial results
Design and power a clinical trial
Learn how the data behave

Limitations

- This study analysed the use of anonymized patient data by cTAP and collaborators to accelerate clinical development
- The study did not include studies directed at accelerating clinical development of new therapeutics that *may* have been conducted by other research groups

Conclusions

- Analysis of natural history and real-world clinical patient data can inform a wide range of important decisions drug developers need to make
- This study, which categorized patient data analyses that help accelerate drug development in DMD, may serve as a template to encourage publication and discussion other groups' work supporting clinical development in rare disease

Disclosures

This study was conducted within the collaborative Trajectory Analysis Project (cTAP), a precompetitive coalition of academic clinicians, drug developers, and patient foundations formed in 2015 to overcome the challenges of high variation in clinical trials in DMD. cTAP has received sponsorship from Astellas (Mitobridge), Avidity Biosciences, BioMarin Pharmaceutical, Bristol Meyers Squibb, Catabasis, Daiichi Sankyo, Edgewise Therapeutics, Entrada Therapeutics, FibroGen, Italfarmaco SpA, Marathon Pharmaceuticals, NS Pharma, Pfizer, PTC Therapeutics, Roche, Sarepta Therapeutics, Shire, Solid Biosciences, Summit Therapeutics, Ultragenyx, Vertex Pharmaceuticals, Parent Project Muscular Dystrophy, Charley's Fund, and CureDuchenne, a founding patient advocacy partner and provider of initial seed funding to cTAP.

Presented at the 27th International Hybrid Annual Congress of the World Muscle Society, October 11–15, 2022, Halifax, Nova Scotia, Canada